BEHAVIORAL AND ELECTROPHYSIOLOGICAL CHARACTERIZATION OF ANTIPSYCHOTIC TREATMENTS IN A RODENT MODEL OF PARKINSON'S DISEASE PSYCHOSIS

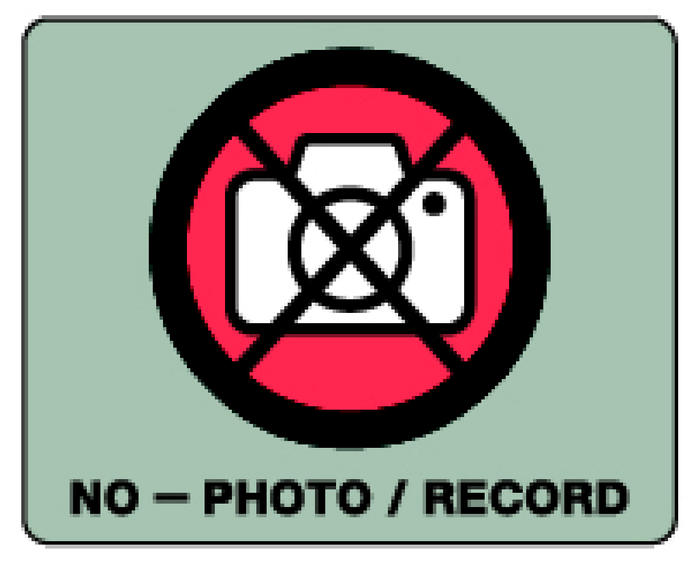
NO – PHOTO / RECORD

T. L. Stan[1], A. Ronaghi[2], S. Barrientos[1], P. Halje[1], **L. Censoni**[2], E. G. Martínez[2], E. Malinina[2], K. Sahlholm[2], P. Petersson[1,2]

[1]*Lund Univ., Lund, Sweden* [2]*Umeå Univ., Umeå, Sweden*

Poster #
449.20

INTRODUCTION

The detection of high frequency oscillations (HFOs) in certain cognitive/limbic structures (such as the Nucleus Accumbens) has previously been recognized as a marker of neuropsychiatric conditions such as schizophrenia. Recently, HFOs have been observed after administration of both classical and dissociative psychedelics as well.

Simultaneously, until somewhat recently, non-motor symptoms of Parkinson's Disease, such as psychosis, visual hallucinations, and auditory hallucinations, have not received a lot of attention, with e. g. a lack of consensus in the literature as to whether they comprise primary symptoms or treatment side effects.

Here, we investigated whether there exists a sensitization of cognitive/limbic structures to hypersynchronous states in Parkinson's Disease, leading to the establishment of HFOs in association with non-motor symptoms, as a possible counterpart to the known effects in motor structures, such as narrowband gamma oscillations, that have been associated with involuntary movements and dyskinesia.

EXPERIMENTAL PROTOCOL

N=8 adult female Sprague-Dawley rats were part of study. National and local guidelines for animal welfare were followed and all procedures were approved in advance by the *Swedish Ethical Committee for Northern Sweden* (Dnr. A15-2018) and by the *Malmö/Lund ethical committee of animal experiments* (Ethic permit number 9/18 01689/2019). Animals were rendered hemiparkinsonian via unilateral 6-OHDA lesions of the medial forebrain bundle.

Experiments consisted of simultaneous electrophysiological and behavioral recordings in awake and freely-behaving animals chronically implanted with multi-electrode arrays. The arrays targeted several structures in the cortico-thalamic-basal ganglia loop, and consisted of 64 recording electrodes per hemisphere. A low-dose NMDA antagonist (MK801) was administered to induce a psychosis-like state, against which 4 distinct treatments were evaluated: Clozapine, Pimavanserin, Mesdopetam and SB277011.

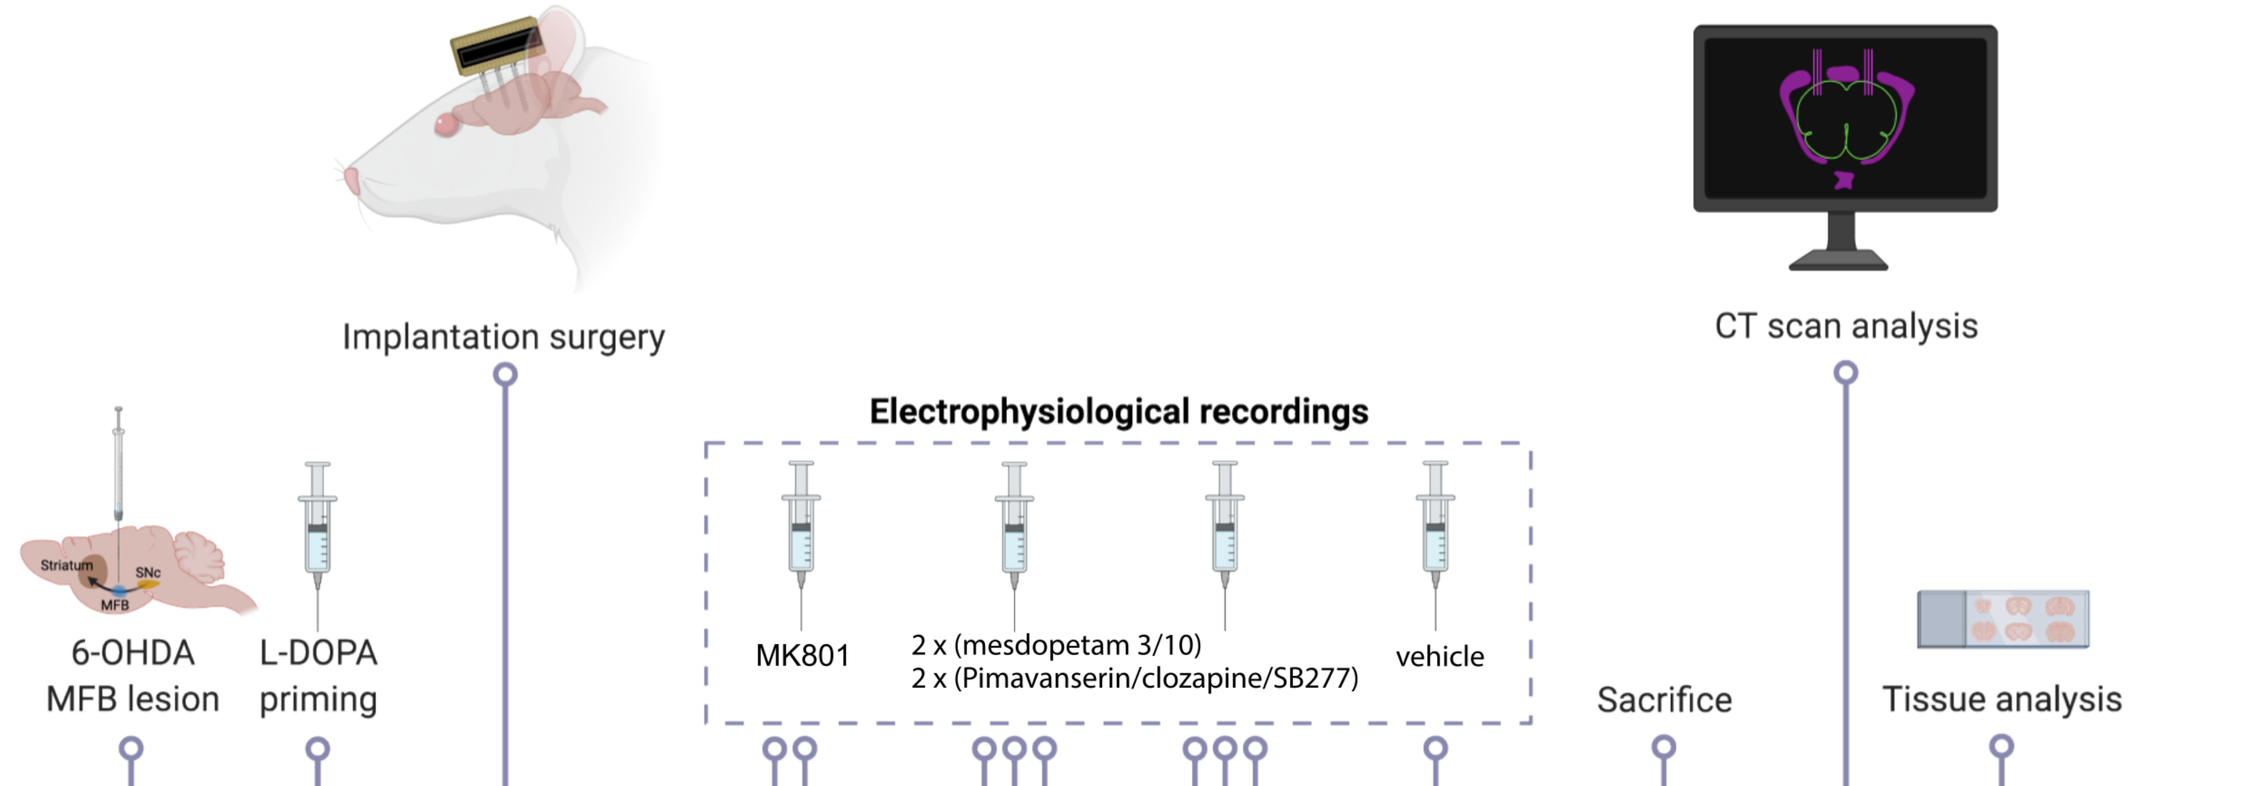
Implantation surgery
CT scan analysis
Electrophysiological recordings
6-OHDA
MFB lesion
L-DOPA
priming
MK801
2 x (mesdopetam 3/10)
2 x (Pimavanserin/clozapine/SB277)
vehicle
Sacrifice
Tissue analysis

DRUG	DOSE (mg/kg)	ADMINISTRATION ROUTE	ADMINISTRATION TIME
MK801	0.05- 0.07	i.p	approx. 20 min after baseline
Clozapine	5	s.c.	30´ before MK801
SB277011A	0.3	s.c.	30´ before MK801
mesdopetam	10	s.c.	Co-administration MK801
mesdopetam	3	s.c.	Co-administration MK801
Pimavanserin	3	s.c.	60´ before MK801

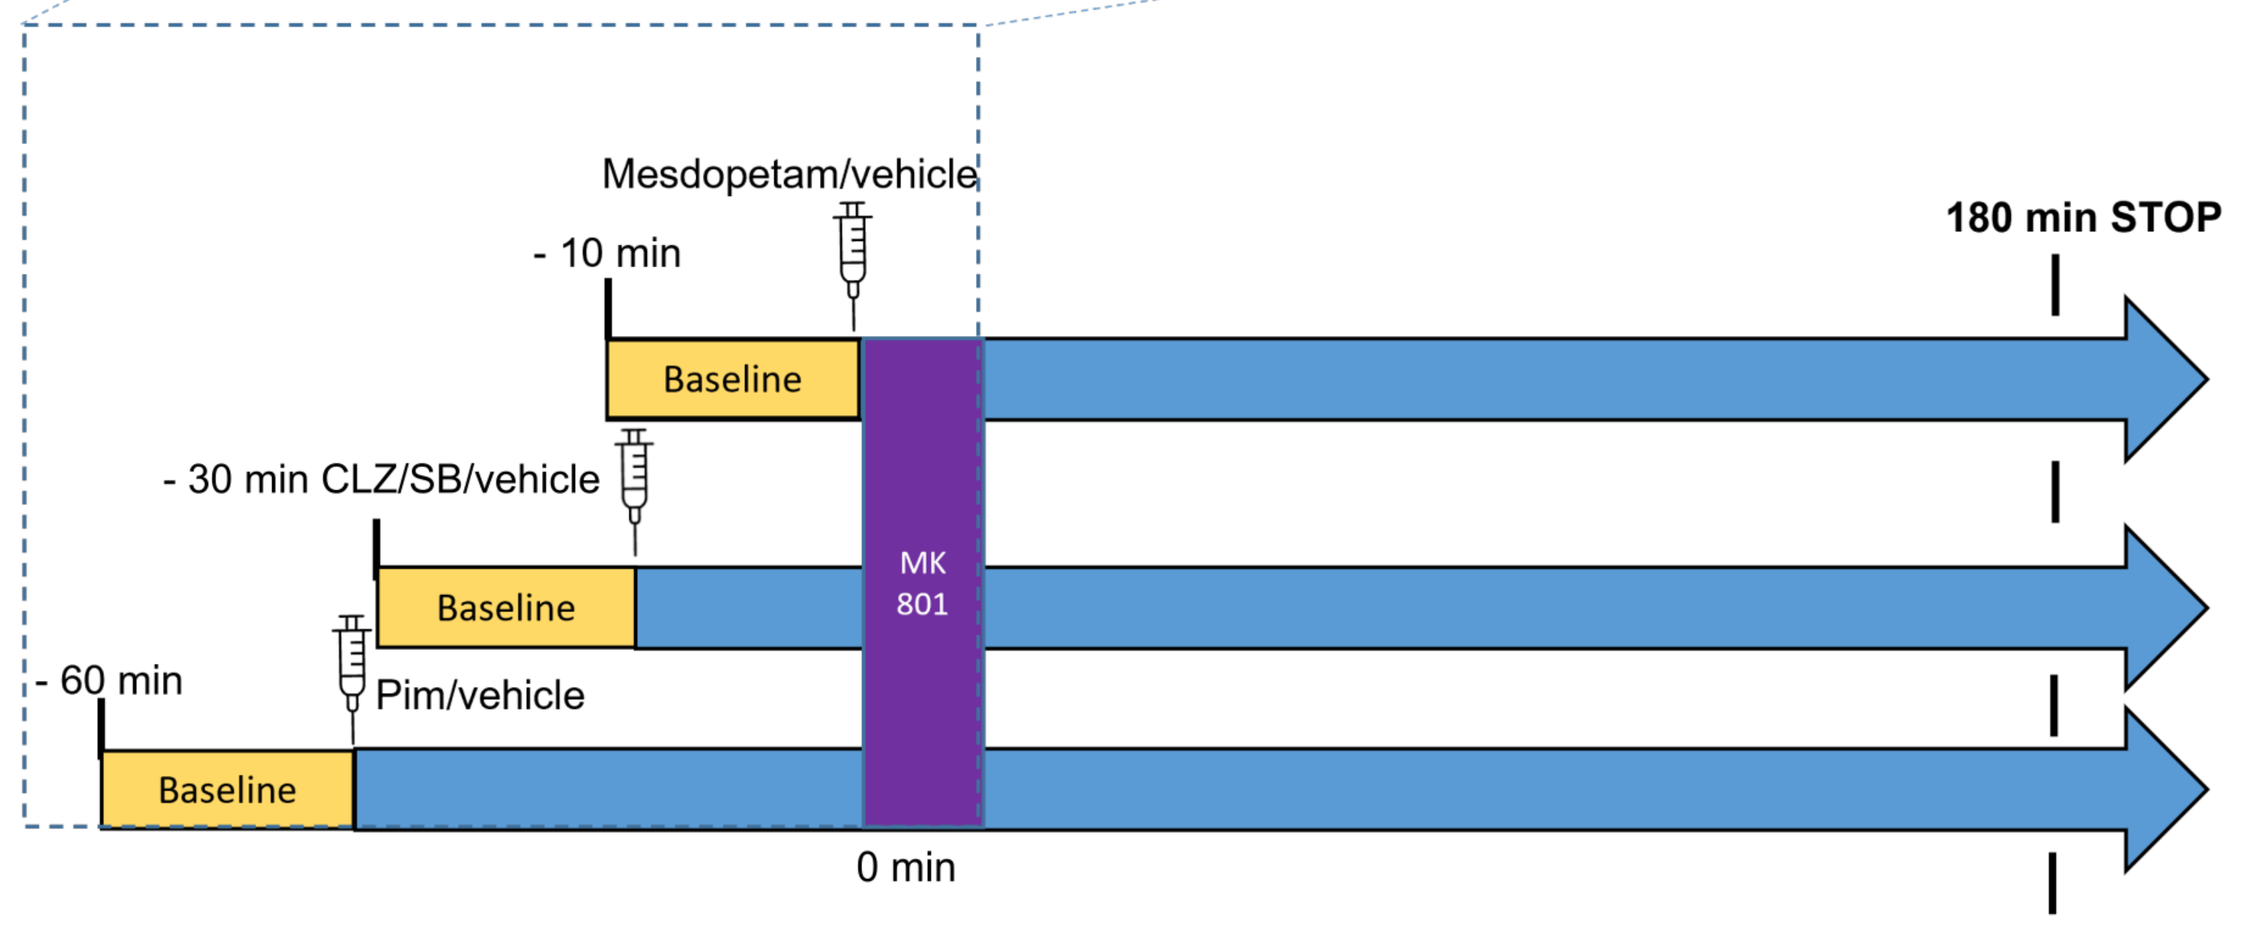
Mesdopetam/vehicle
- 10 min
180 min STOP
Baseline
- 30 min CLZ/SB/vehicle
Baseline
MK 801
- 60 min
Pim/vehicle
Baseline
0 min

METHODS

Behavior during experiments was recorded with video cameras and analyzed offline. Body parts were tracked using *DeepLabCut*, and behavioral features were quantified with purpose-made algorithms.

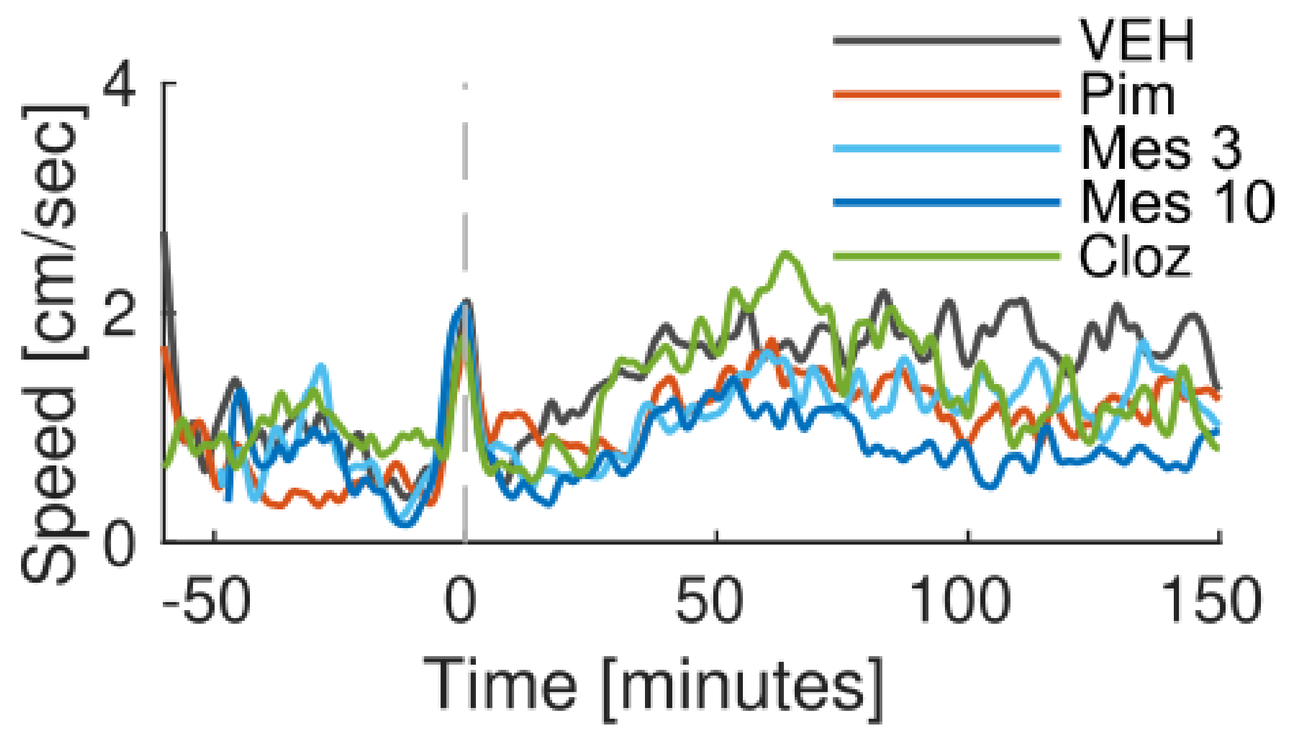
VEH
Pim
Mes 3
Mes 10
Cloz
Speed [cm/sec]
Time [minutes]

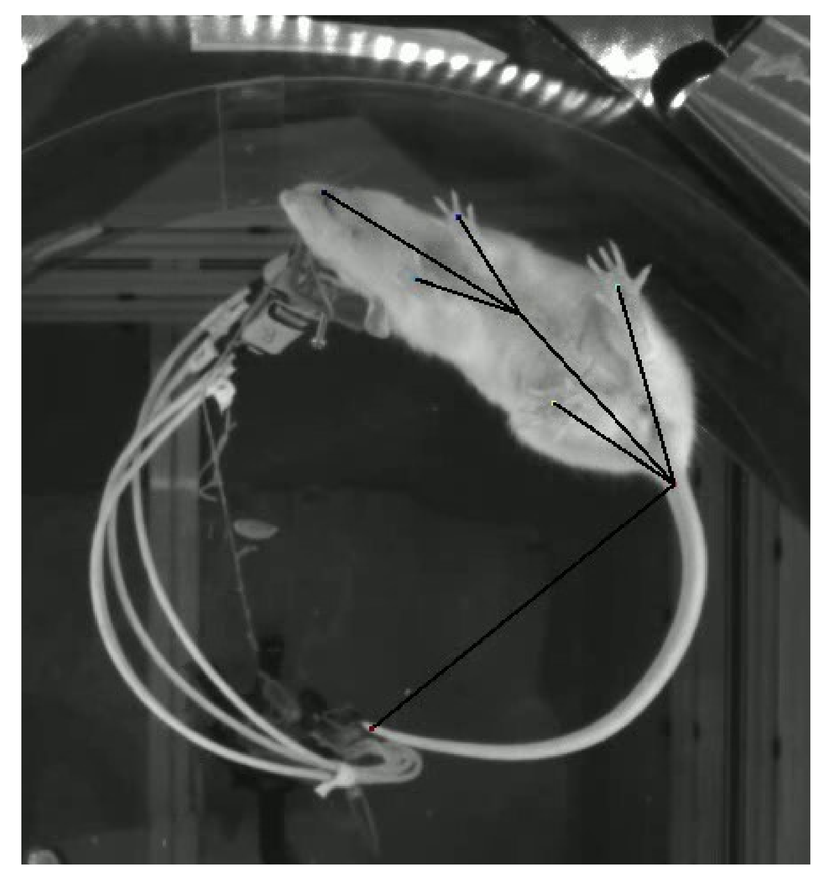

The position of recorded electrodes was verified *post mortem,* through CT images analyzed according to the method described in (Censoni *et al.*, 2022). Wires were assigned anatomical tags and subsequently functionally grouped into larger structures.

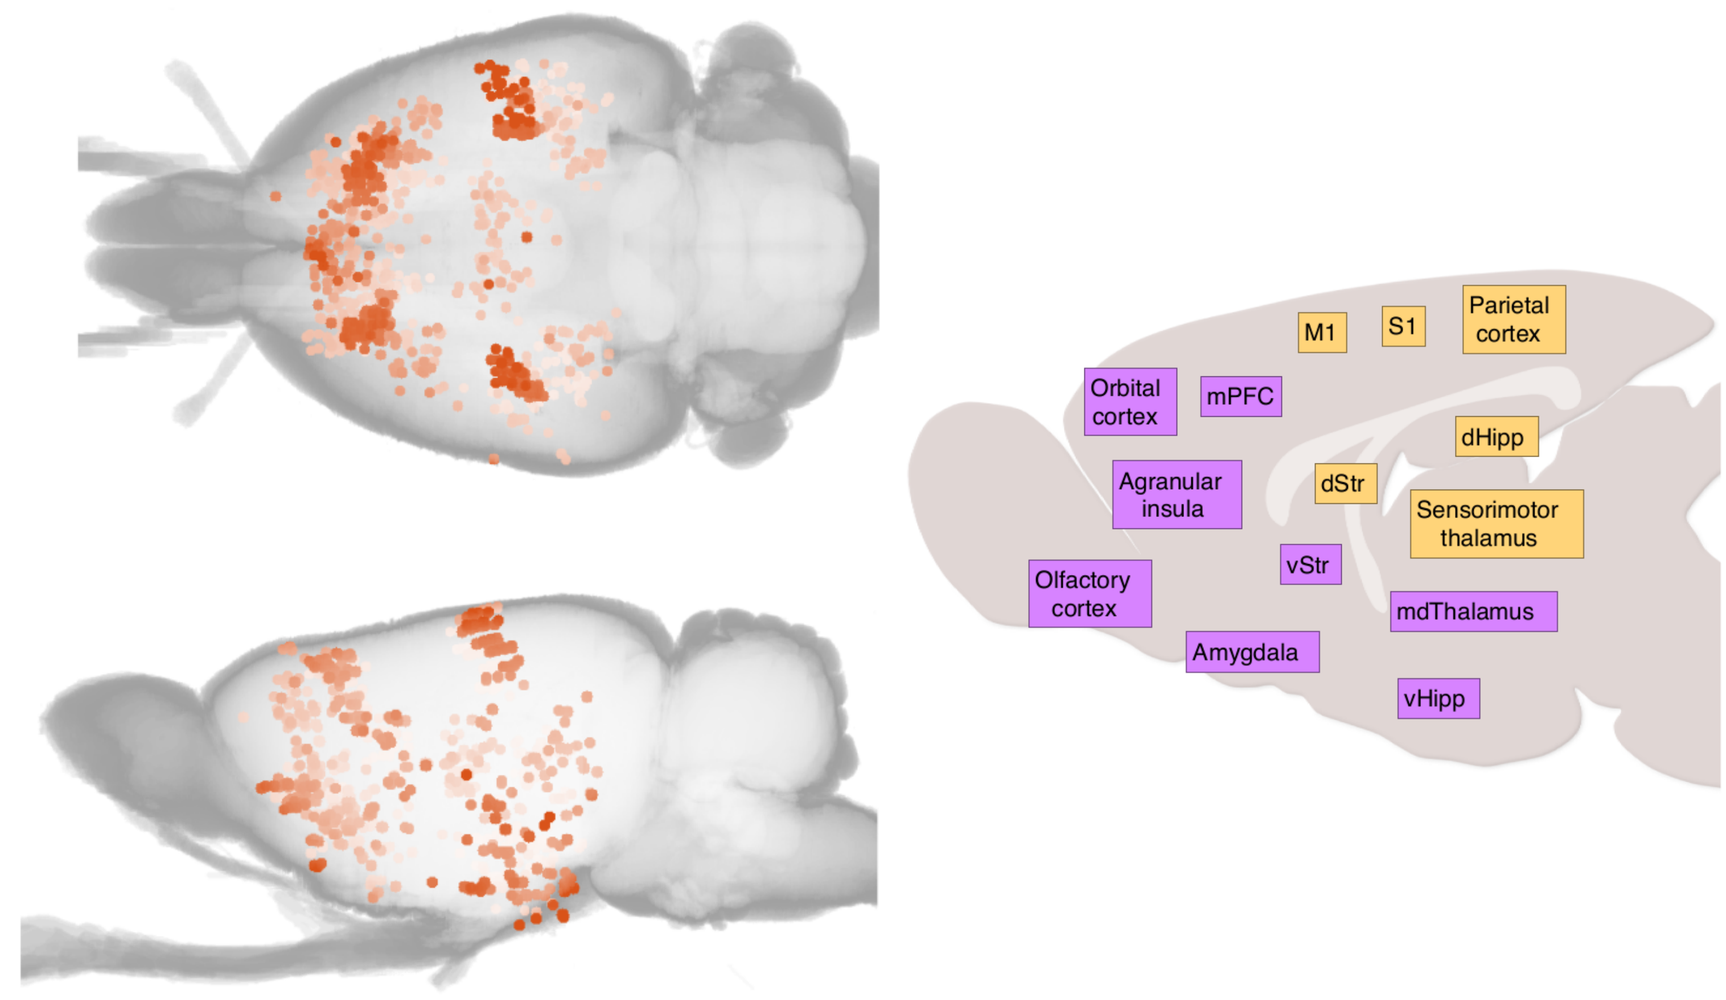
M1
S1
Parietal cortex
Orbital cortex
mPFC
dHipp
Agranular insula
dStr
Sensorimotor thalamus
vStr
Olfactory cortex
mdThalamus
Amygdala
vHipp

Number of sessions, Intact hemisphere

	base	MK801+saline	MK801+DMSO	MK801+mesdopetam 3mg	MK801+mesdopetam 10mg	MK801+pimavanserin	MK801+clozapine	MK801+SB277011
olf	105	9	3	11	11	11	13	7
AI	84	5	5	8	8	8	8	6
OFC	108	8	2	9	9	9	11	7
PFC	80	9	2	9	9	9	11	6
StrV	82	9	2	9	9	9	11	6
amyg	103	9	4	10	10	10	12	8
hippV	130	9	4	11	11	11	13	9
Cl	5	0	0	1	1	1	1	0
MD	61	4	2	5	5	5	7	3
M1	56	5	0	5	5	5	7	3
S1	125	7	5	10	10	10	12	7
PtA	68	5	2	6	6	6	6	6
StrD	113	10	4	11	11	11	13	8
hippD	8	1	0	1	1	1	1	0
thal	99	6	6	8	8	8	10	6

Number of sessions, Lesioned hemisphere

	base	MK801+saline	MK801+DMSO	MK801+mesdopetam 3mg	MK801+mesdopetam 10mg	MK801+pimavanserin	MK801+clozapine	MK801+SB277011
olf	52	3	2	4	4	3	5	3
AI	29	1	2	2	2	2	2	2
OFC	135	9	4	11	11	11	13	9
PFC	79	7	0	7	7	7	9	5
StrV	108	10	4	11	11	11	13	8
amyg	80	9	2	9	9	9	11	6
hippV	79	5	6	7	7	7	7	5
Cl	68	5	2	6	6	6	8	5
MD	6	1	0	1	1	1	1	0
M1	130	8	4	10	10	10	12	9
S1	73	4	4	6	6	6	8	4
PtA	83	8	4	9	9	9	9	7
StrD	110	7	6	9	9	9	9	7
hippD	64	2	4	4	4	4	4	4
thal	74	5	4	6	5	6	8	4

Bipolar LFP time series were computed offline from pairs of electrodes located in the same structure, in order to prioritize local sources. To emphasize oscillatory components in the power spectrum, we employed a recent method based on irregular resampling (Wen and Liu, 2016). To isolate arrhythmic components from the power spectrum, the time series were resampled multiple times. Then, by normalizing against the fractal component, we constructed a power spectrum measure that highlights rhythmic activity.

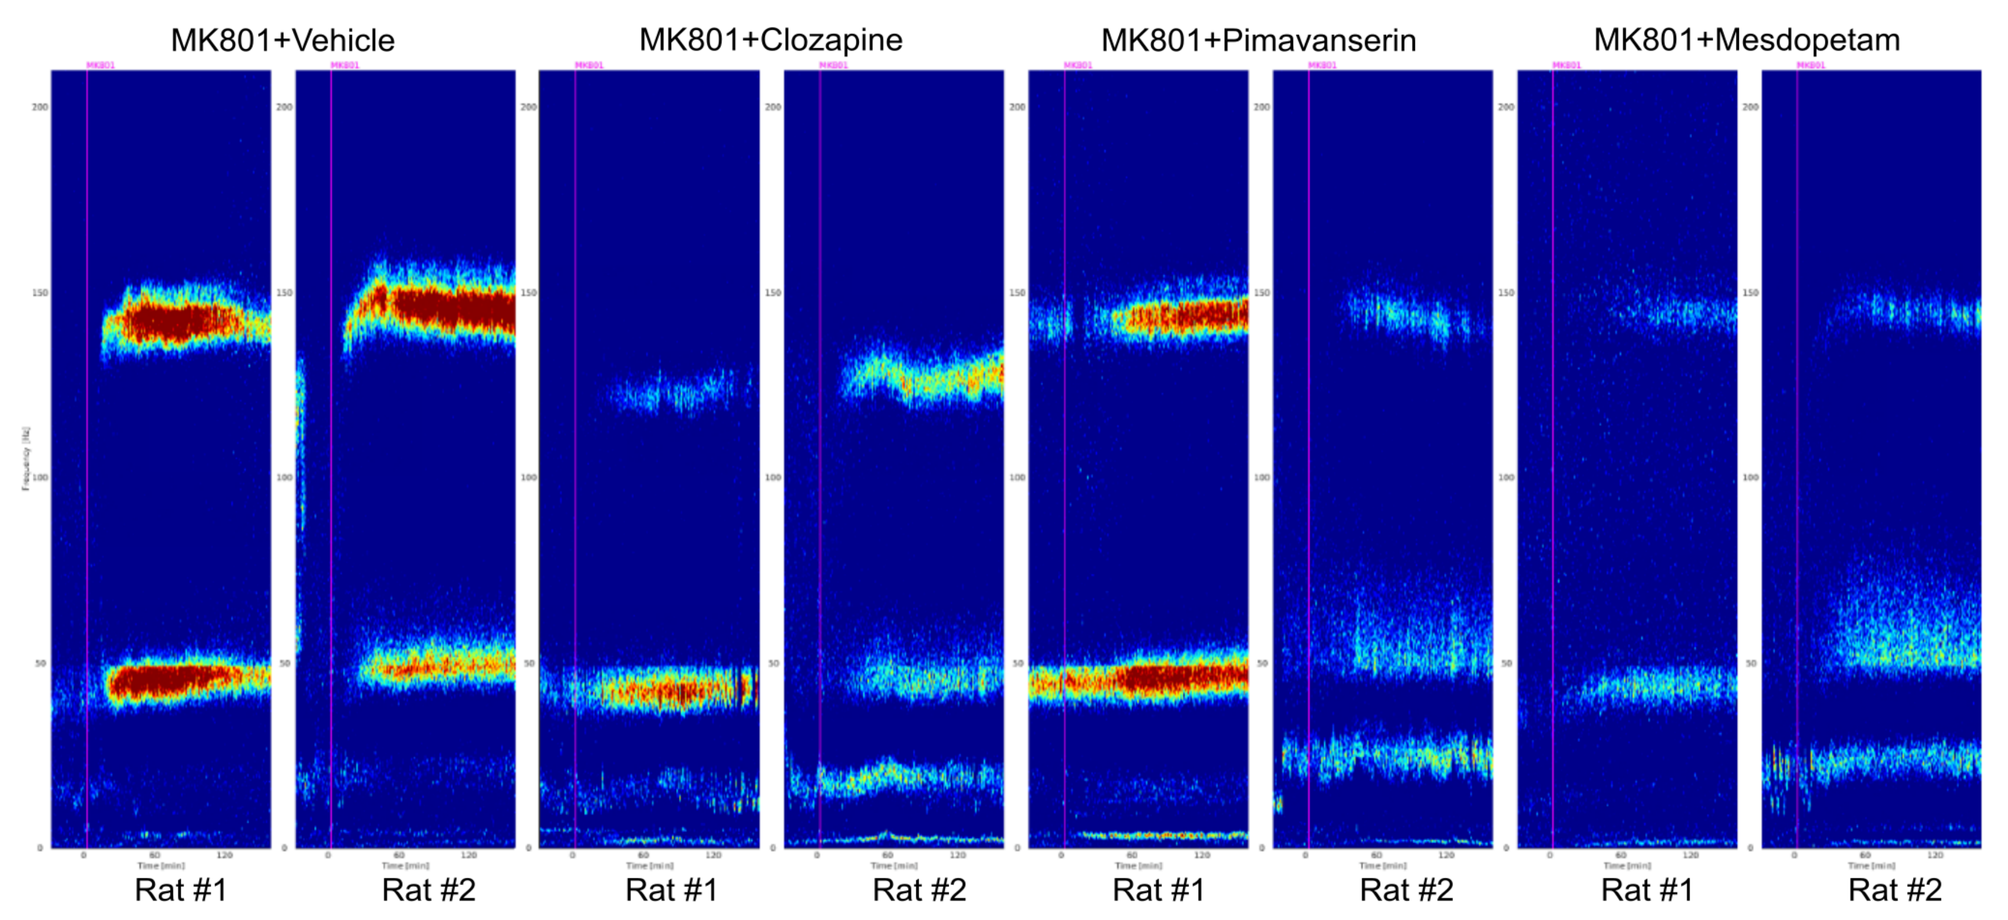
MK801+Vehicle
MK801+Clozapine
MK801+Pimavanserin
MK801+Mesdopetam
Rat #1
Rat #2
Rat #1
Rat #2
Rat #1
Rat #2
Rat #1
Rat #2

Example spectrograms from mPFC in lesioned hemisphere showing HFOs.

RESULTS

The administration of low-dose MK801 produced behavioral effects: increased bout frequency and a tendency for increase in circularly biased motion. All the anti-psychotics tested showed a tendency to normalize bout frequency, and to reduce circularly biased motion. Together, the induced hyperactivity and side bias indicate that the low dose NMDA antagonist is able to induce a psychosis-like state, and that the lesioned hemisphere is likely to be more sensitive compared to the control hemisphere.

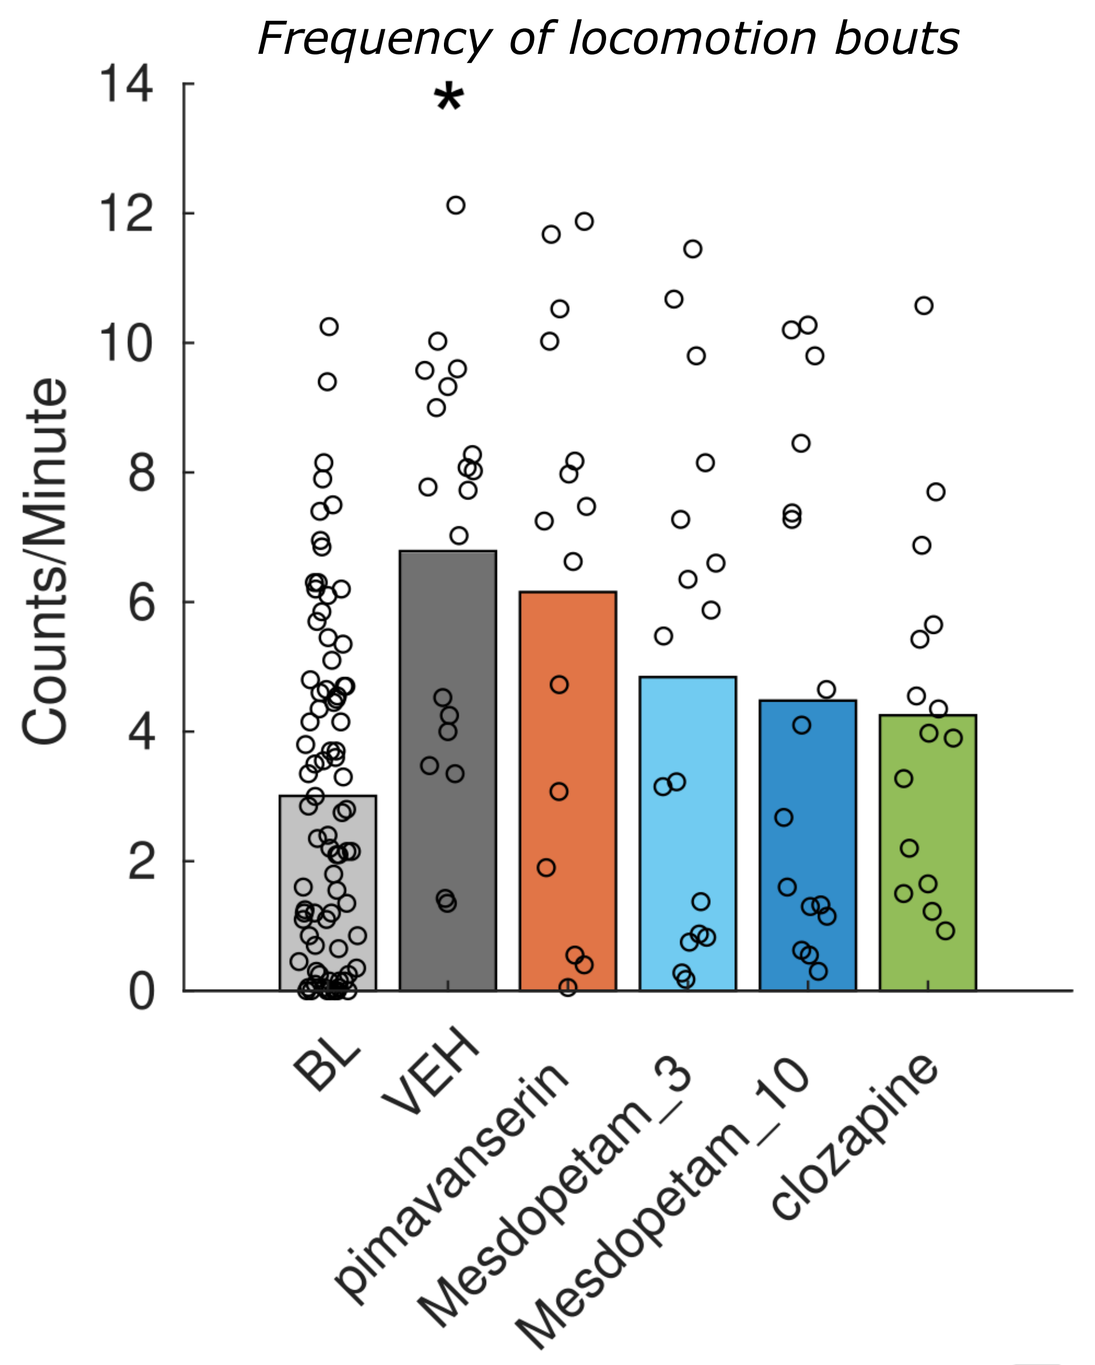
Frequency of locomotion bouts
Counts/Minute
BL
VEH
pimavanserin
Mesdopetam_3
Mesdopetam_10
clozapine

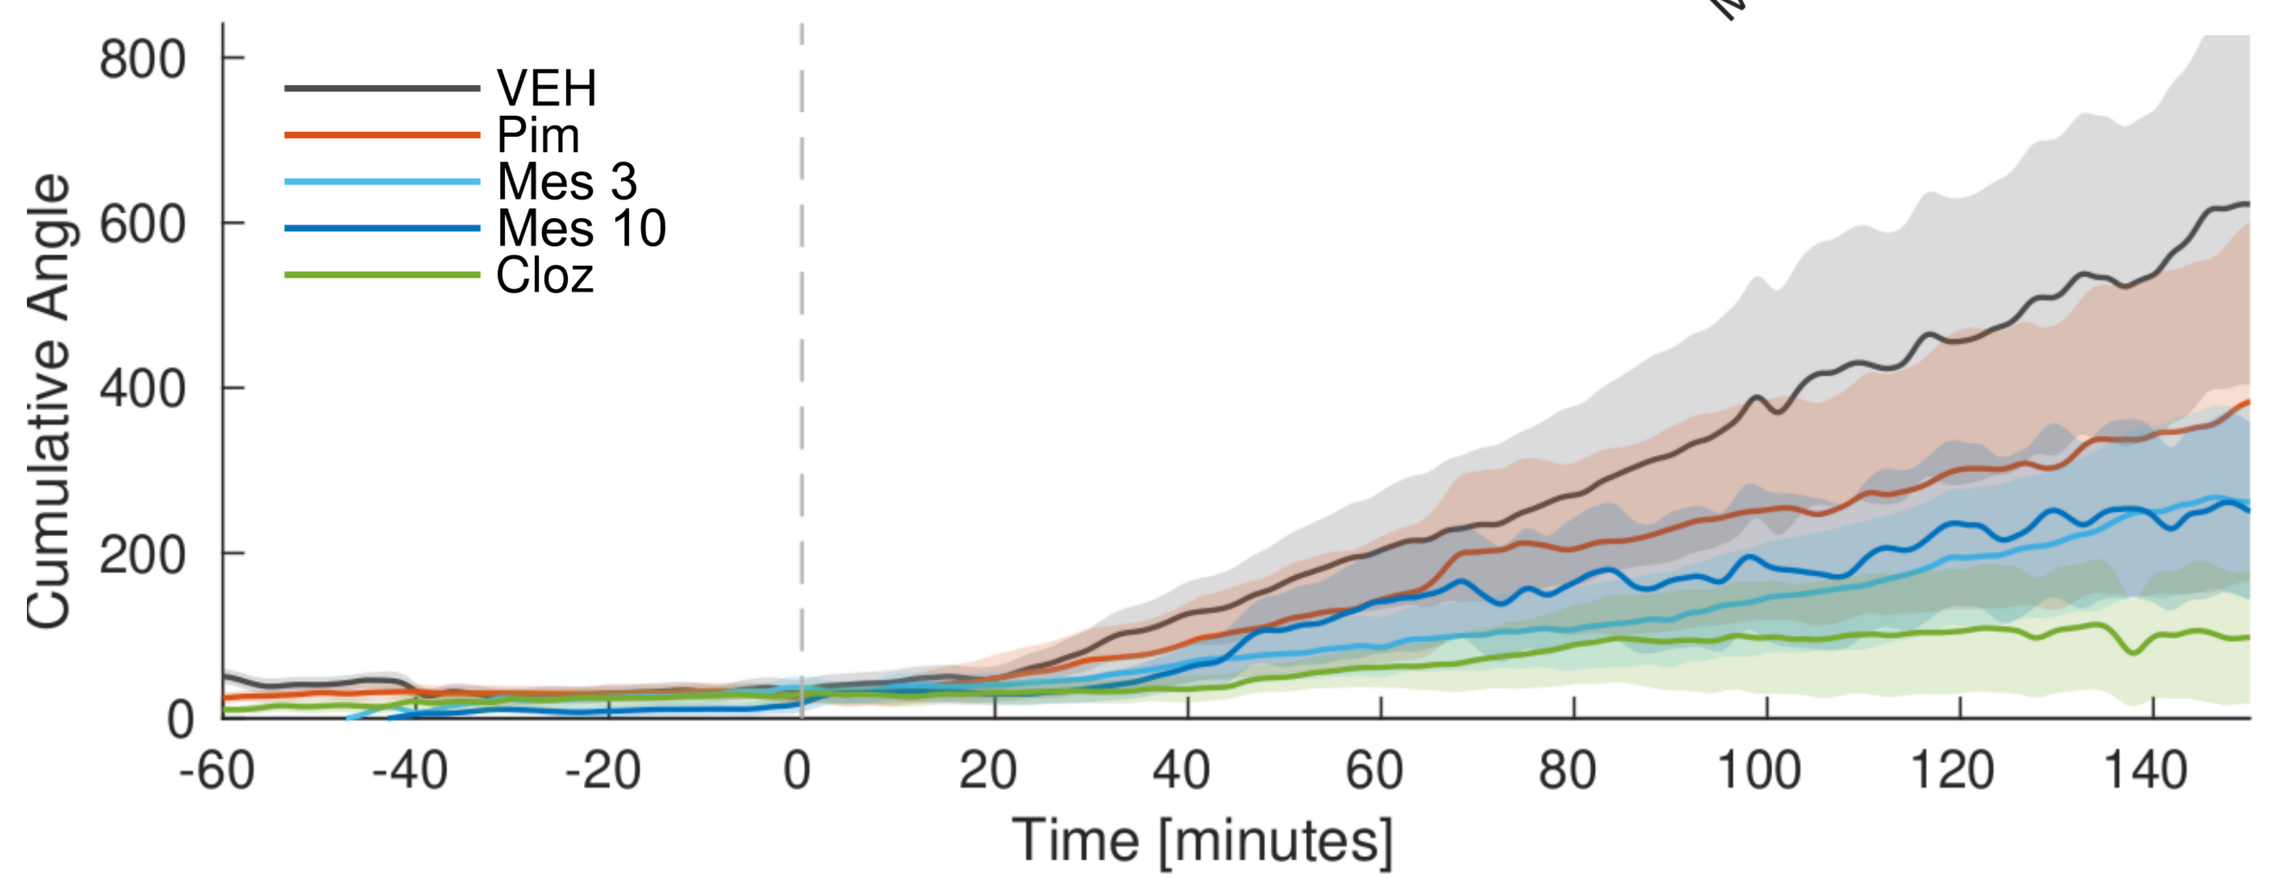
VEH
Pim
Mes 3
Mes 10
Cloz
Cumulative Angle
Time [minutes]

HFOs were detected, albeit at low rates, already during baseline in both hemispheres – but more in the lesioned hemisphere. Non-motor symptoms of Parkinson's Disease typically develop over time, and they are not easy to characterize in animal models. Nevertheless, these detections suggest that this model is already prone to some fashion of non-motor symptoms even before the administration of MK801, and that cognitive/limbic structures in the lesioned hemisphere can sustain hypersynchronous states.

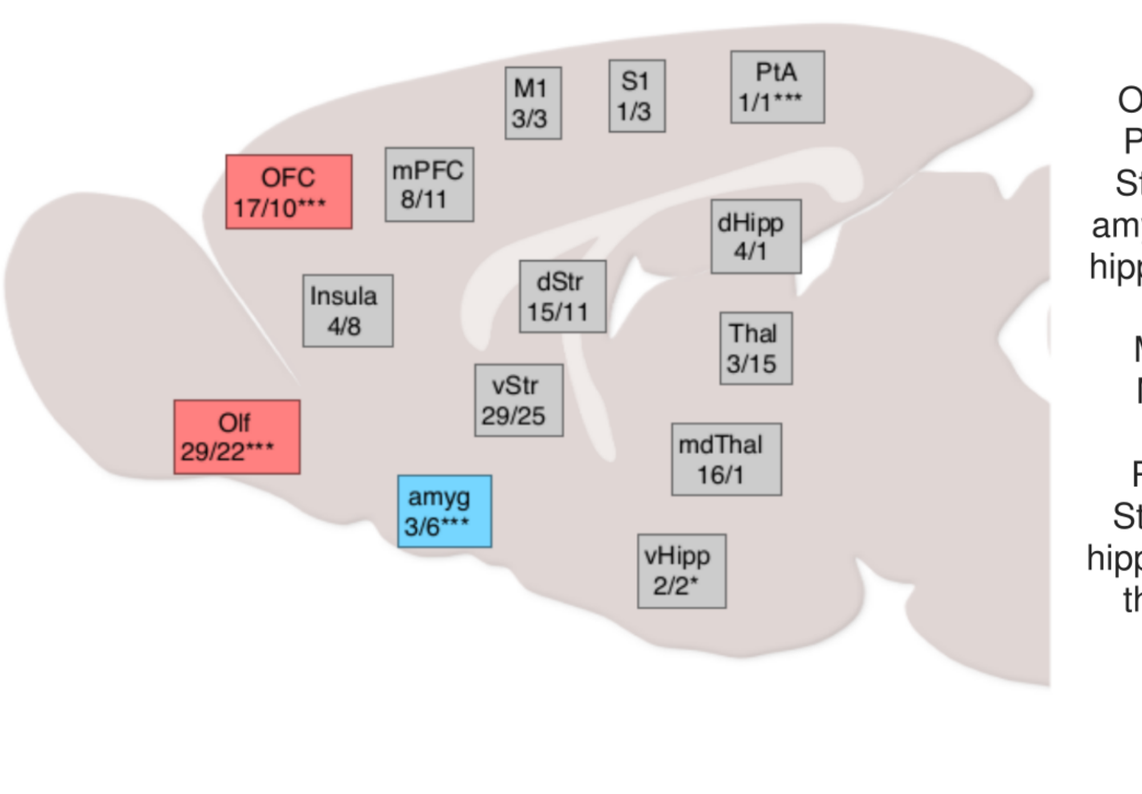
M1 3/3
S1 1/3
PtA 1/1***
OFC 17/10***
mPFC 8/11
dHipp 4/1
Insula 4/8
dStr 15/11
Thal 3/15
vStr 29/25
Olf 29/22***
mdThal 16/1
amyg 3/6***
vHipp 2/2*

	Intact hemisphere: base	Intact hemisphere: MK801+saline	Intact hemisphere: MK801+DMSO	Lesioned hemisphere: base	Lesioned hemisphere: MK801+saline	Lesioned hemisphere: MK801+DMSO
olf	0.04	0.03	0.09	0.47	0.18	0.24
AI	0.04	0.04	0.03	0.06	0.04	0.03
OFC	0.20	0.34	0.49	0.35	0.19	0.22
PFC	0.12	0.33	0.28	0.14	0.70	
StrV	0.33	0.24	0.27	0.40	0.29	0.29
amyg	0.02	0.02	0.05	0.04	0.01	0.01
hippV	0.01	0.03	0.02	0.08	0.15	0.07
Cl	0.04			0.22	0.04	0.27
MD	0.03	0.06	0.08	0.20	0.08	
M1	0.03	0.15		0.02	0.11	0.01
S1	0.04	0.13	0.16	0.02	0.21	0.30
PtA	0.06	0.09	0.23	0.02	0.05	0.02
StrD	0.09	0.06	0.15	0.04	0.03	0.05
hippD	0.03	0.03		0.01	0.01	0.06
thal	0.03	0.06	0.05	0.02	0.09	0.03

HFO detection rates. Left: lesioned vs. intact hemisphere during baseline. Right: rates in all structures during baseline and MK801 state.

Moreover, the administration of MK801 induces a pattern of widespread HFOs that reaches cognitive/limbic structures in both hemispheres.

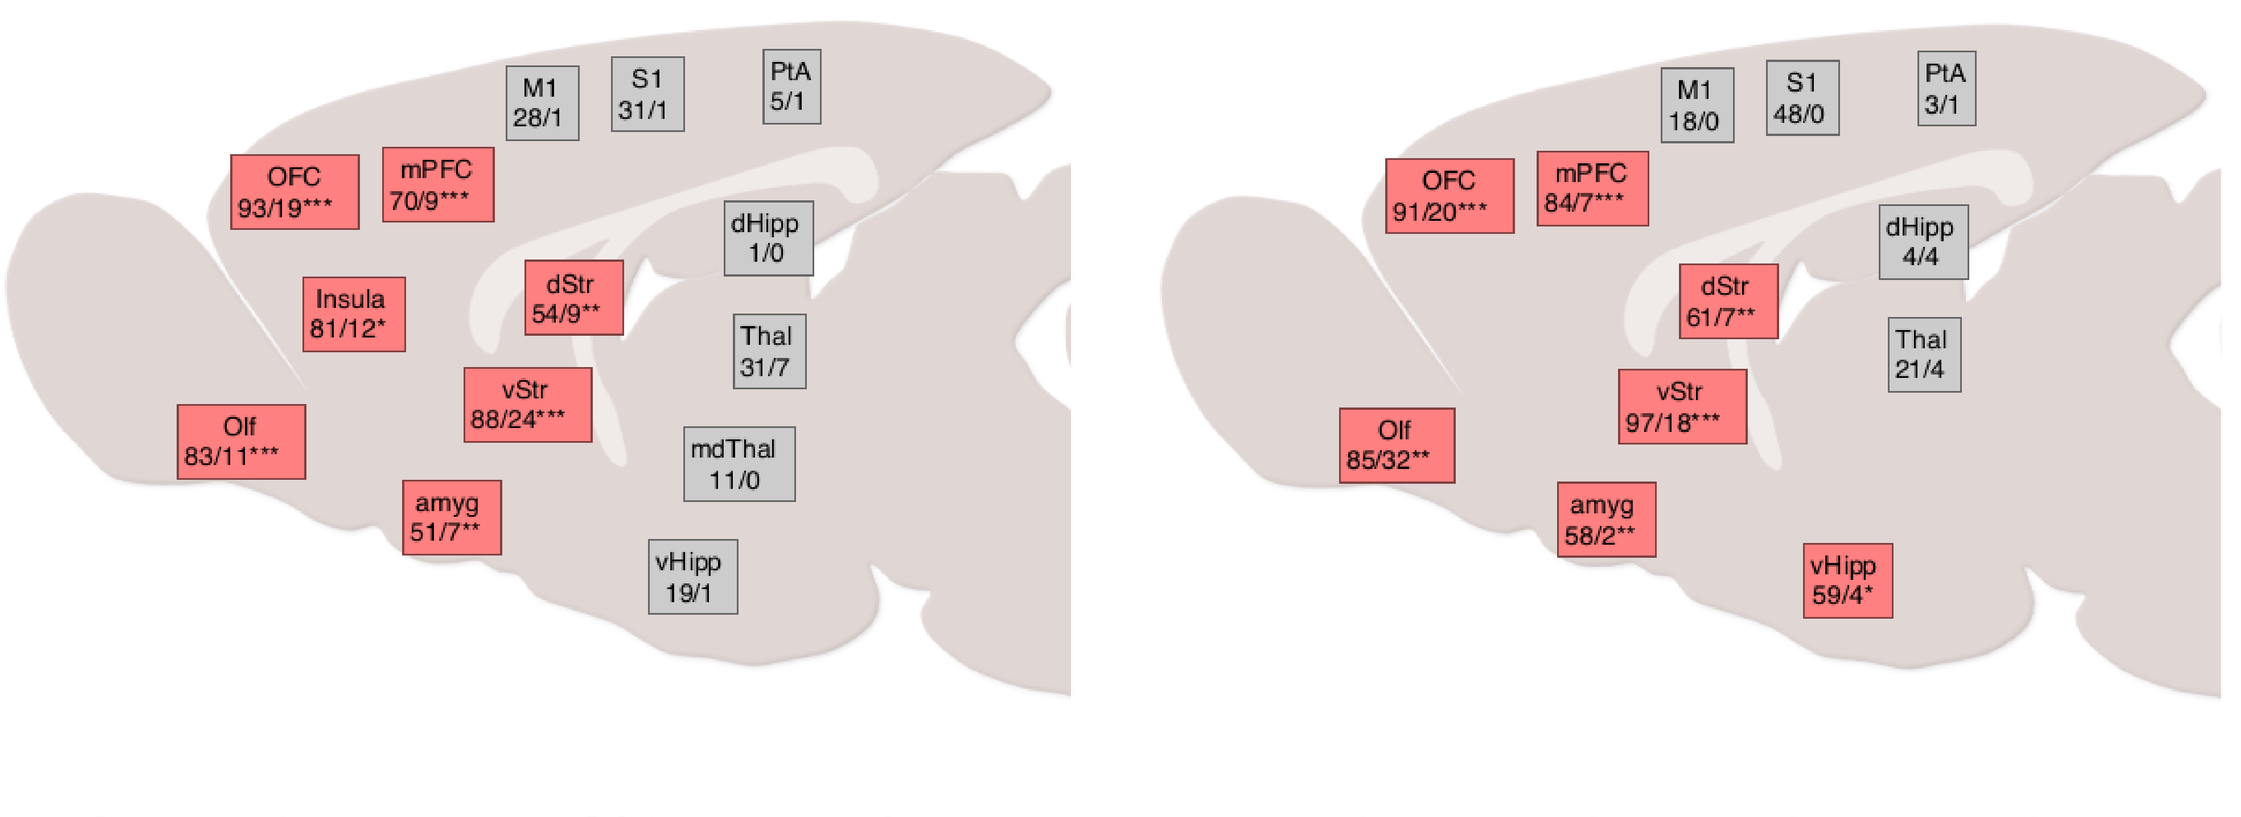
M1 28/1
S1 31/1
PtA 5/1
OFC 93/19***
mPFC 70/9***
dHipp 1/0
Insula 81/12*
dStr 54/9**
Thal 31/7
vStr 88/24***
Olf 83/11***
mdThal 11/0
amyg 51/7**
vHipp 19/1
M1 18/0
S1 48/0
PtA 3/1
OFC 91/20***
mPFC 84/7***
dHipp 4/4
dStr 61/7**
Thal 21/4
vStr 97/18***
Olf 85/32**
amyg 58/2**
vHipp 59/4*

HFO detection rates, MK801 vs. baseline. Left: Intact hemisphere. Right: Lesioned hemisphere.

All the treatments we tested reduced the detection of HFOs to some extent. Nevertheless, different treatments exhibited distinct profiles in terms of the structures affected.
Clozapine was the only treatment observed to reduce HFOs in the ventral striatum and ventral hippocampus, while the effects of Mesdopetam and SB277011 appeared to be more localized to the medial prefrontal cortex. Pimavanserin and Clozapine also modulated the detection of HFOs in dorsal striatum and primary somatosensory cortex.

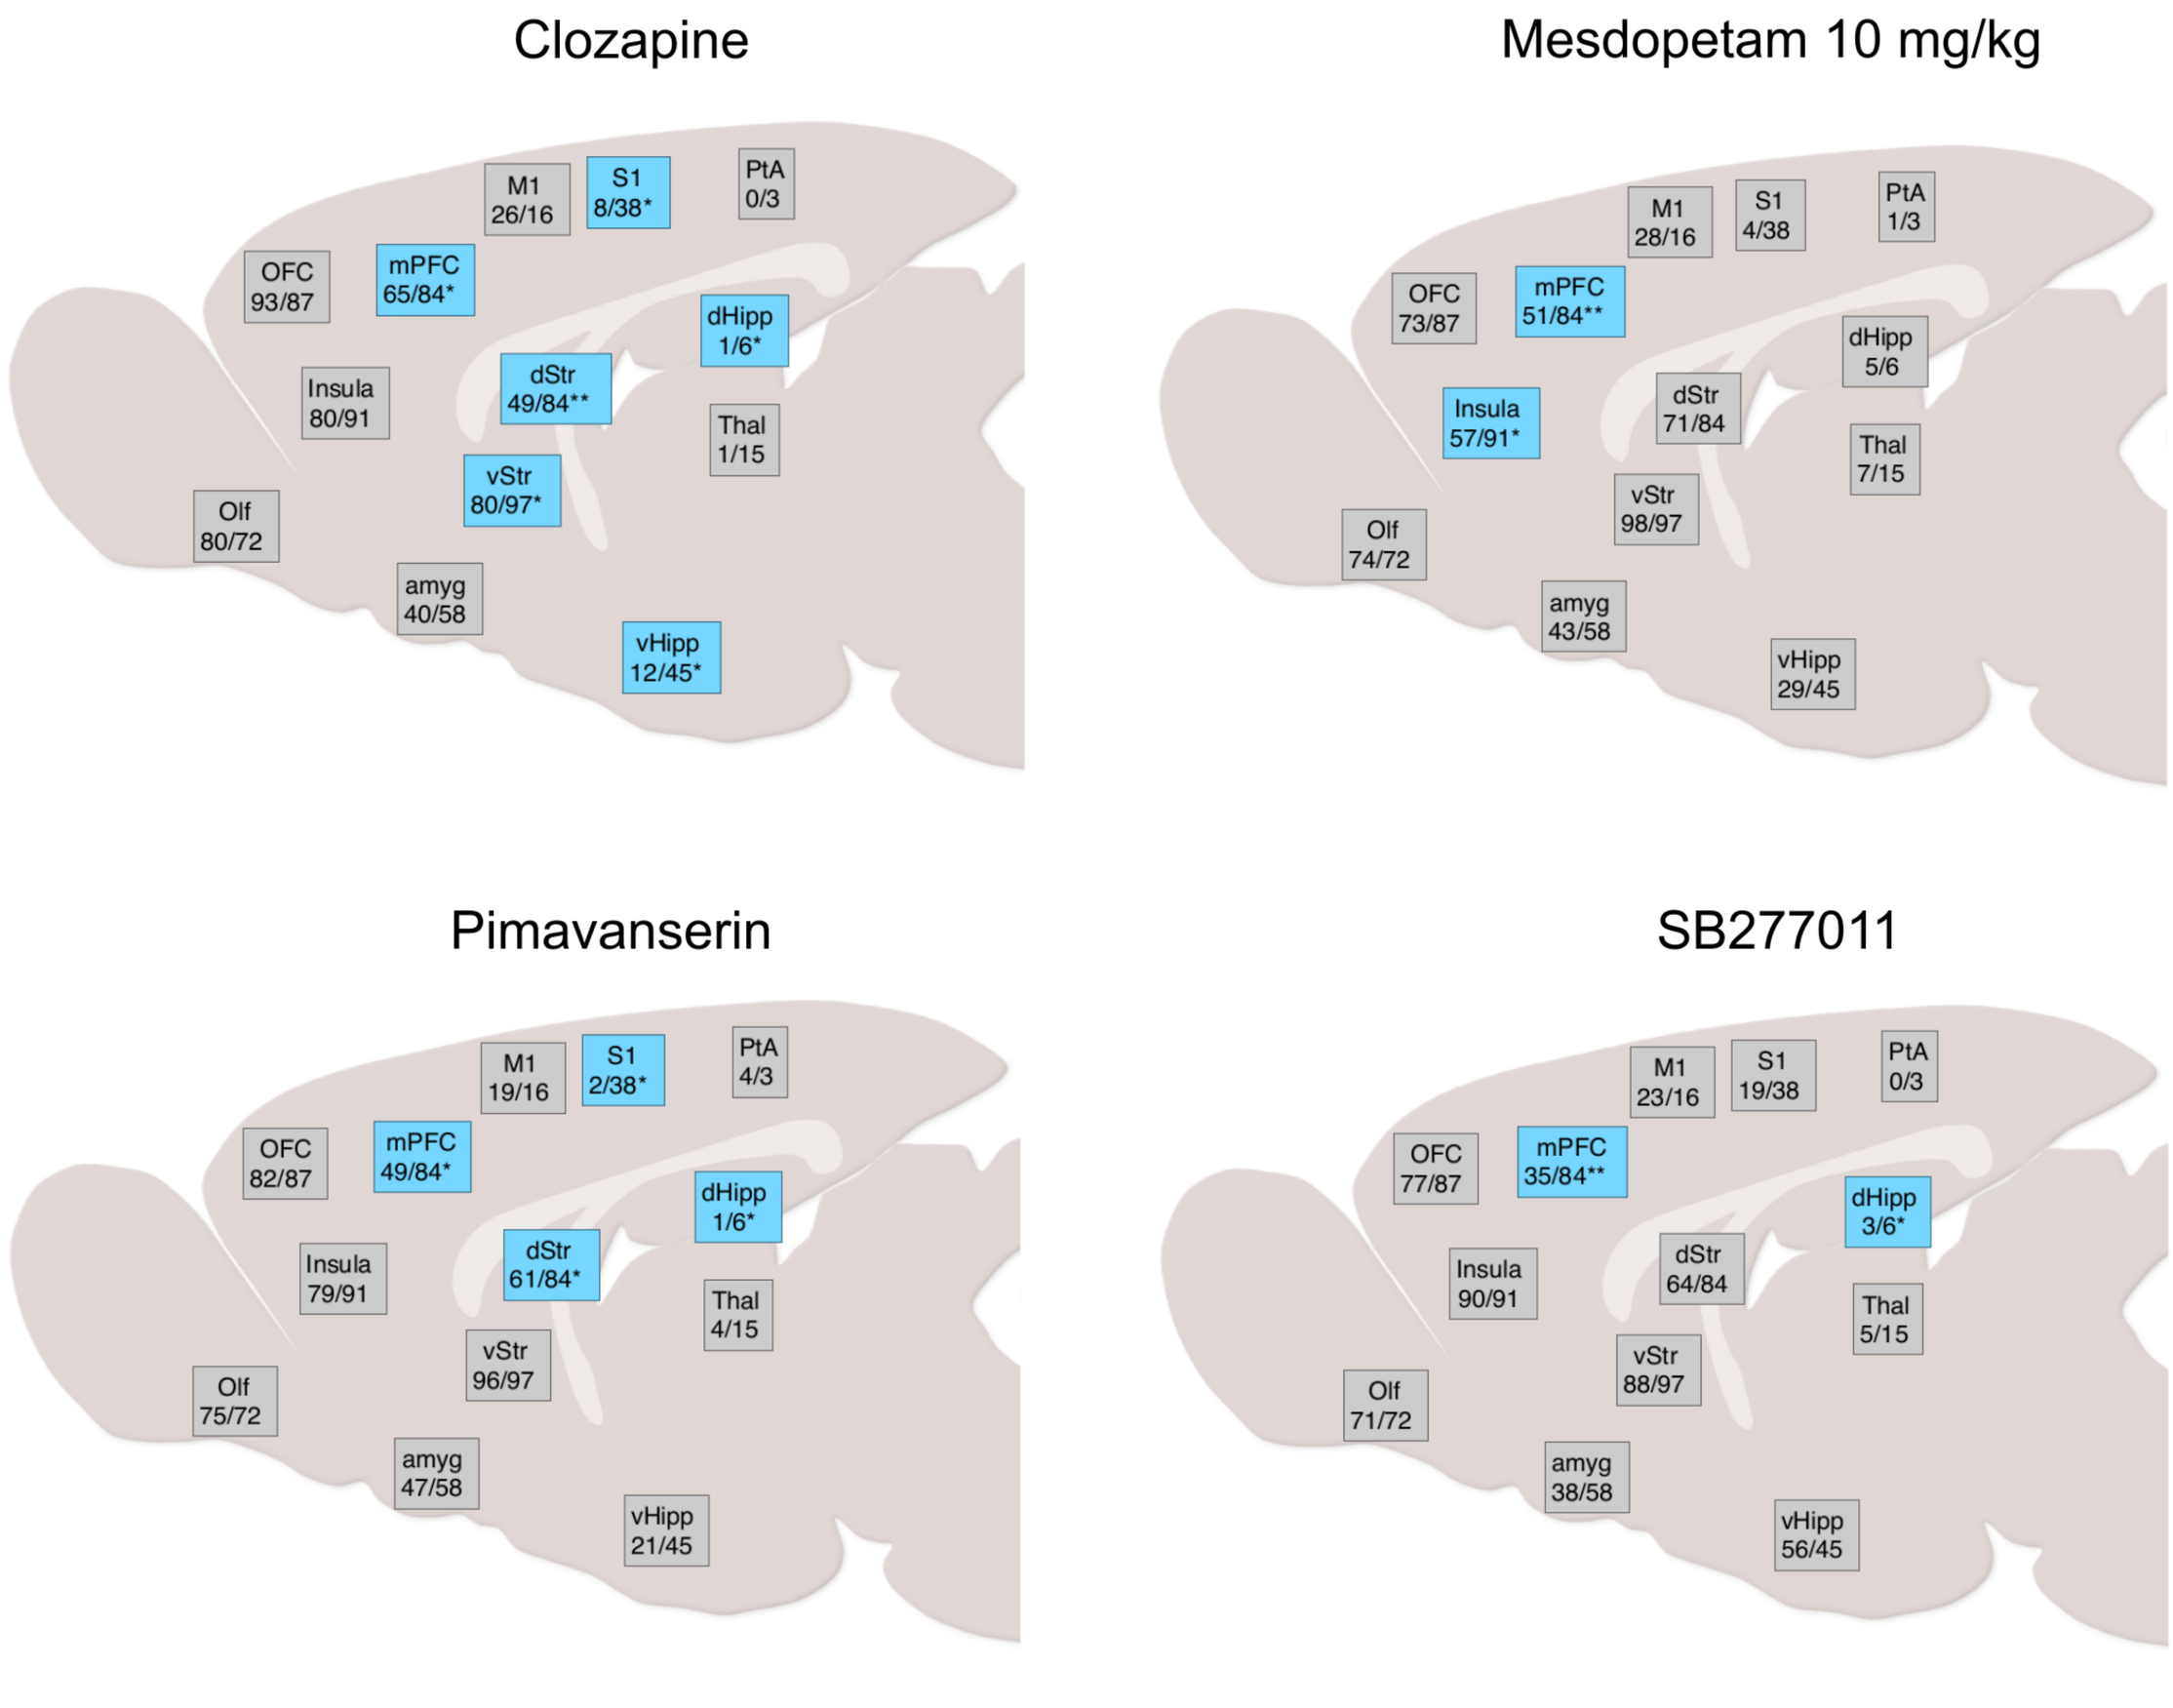
Clozapine
M1 26/16
S1 8/38*
PtA 0/3
OFC 93/87
mPFC 65/84*
dHipp 1/6*
Insula 80/91
dStr 49/84**
Thal 1/15
vStr 80/97*
Olf 80/72
amyg 40/58
vHipp 12/45*
Mesdopetam 10 mg/kg
M1 28/16
S1 4/38
PtA 1/3
OFC 73/87
mPFC 51/84**
dHipp 5/6
Insula 57/91*
dStr 71/84
Thal 7/15
vStr 98/97
Olf 74/72
amyg 43/58
vHipp 29/45
Pimavanserin
M1 19/16
S1 2/38*
PtA 4/3
OFC 82/87
mPFC 49/84*
dHipp 1/6*
Insula 79/91
dStr 61/84*
Thal 4/15
vStr 96/97
Olf 75/72
amyg 47/58
vHipp 21/45
SB277011
M1 23/16
S1 19/38
PtA 0/3
OFC 77/87
mPFC 35/84**
dHipp 3/6*
Insula 90/91
dStr 64/84
Thal 5/15
vStr 88/97
Olf 71/72
amyg 38/58
vHipp 56/45

HFO detection rates, treatment vs. MK801, in the lesioned hemisphere.

Additionally, we have performed the same suite of analyses with respect to altered power in the gamma band, and we are looking into modulatory effects on the activity of single units. These results are currently in preparation, but together they will allow us to clarify the differences between the baseline, MK801, and each of the treated states, in the intact and lesioned hemispheres.

CONCLUSIONS

- In Parkinson's Disease, cognitive/limbic structures are able to sustain high frequency local field potential oscillations, and they appear to be associated with psychosis-like states.
- These observations echo the observation of narrowband gamma oscillations in motor structures during dyskinesia.
- These oscillations become significantly more prevalent after the administration of the NMDA antagonist MK801.
- Anti-psychotic treatments are able to modulate the prevalence of high frequency oscillations and the behavioral manifestations simultaneously.
- Treatments with distinct mechanisms of action modulate the detection of high frequency oscillations in different structures.

ACKNOWLEDGEMENTS

This work was partly supported by a grant from the "Swedish Governmental Agency for Innovation Systems – Vinnova"'s program "Swelife and Medtech4Health – Collaboration project for better health spring 2019."

UMEÅ UNIVERSITY

Presenter: Luciano Censoni
Find me at *https://luciano-censoni.github.io/*