Behavioral and electrophysiological characterization of the antidyskinetic treatments in a rodent model of PD-LID

A. Ronaghi[1], T. Loredan Stan[2], S.A. Barrientos[2], S. Sulis-Sato[1], E. Malinina[1], L. Censoni[1], P. Halje[2], P. Petersson[1,2]
1. Integrative Medical Biology, Umeå University, Umeå, Sweden. 2. Integrative Neurophysiology and Neurotechnology, Experimental Medical Science, Lund University, Lund, Sweden

#449.21

UMEÅ UNIVERSITY

Introduction

In Parkinson´s disease (PD), pharmacological treatment with the dopamine precursor levodopa, in the long-term, often leads to troublesome side effects in the form of levodopa-induced dyskinesia (LID). To mitigate these problems, an anti-dyskinetic drug, such as amantadine, may be used together with levodopa treatment. However, the use of amantadine is sometimes limited by other side effects, particularly in the psychiatric domain. We have here investigated how the novel drug candidate, mesdopetam, compares to amantadine and pimavanserin with respect to reduction of LID in a rat model of PD-LID.

Methods

To explore the mechanistic effects of the different drugs on LID, the severity of dyskinesia - quantified by scoring of abnormal involuntary movement (AIMs) - and hyperkinesia - detected by automatic video tracking of body rotations - of hemiparkinsonian rats under different drug administration was analyzed. Simultaneously, local field potentials were recorded to detect physiological changes in brain activity, e.g. narrow band gamma oscillations (NBGs), known to be associated with LID, in motor related and limbic brain structures (in total 128 electrodes, distributed in 17 structures in each hemisphere.

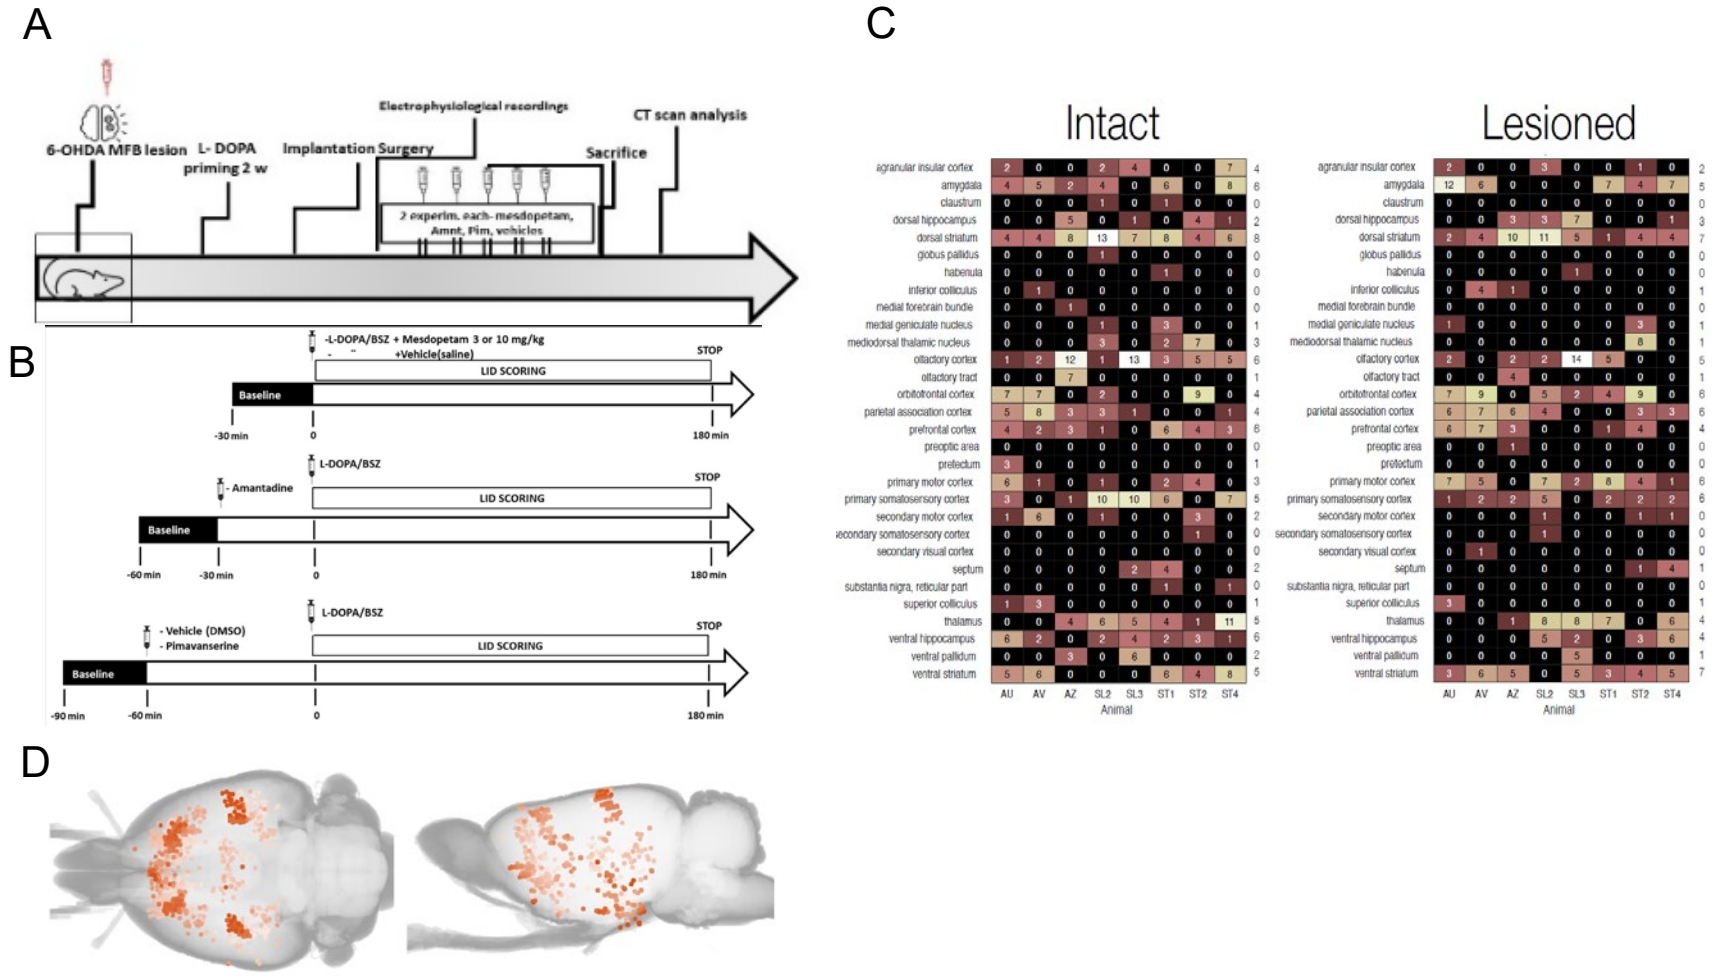
A
C
D
6-OHDA MFB lesion
L-DOPA priming 2 w
Implantation Surgery
Electrophysiological recordings
CT scan analysis
Sacrifice
2 experim. each- mesdopetam, Amnt, Pim, vehicles
B
-L-DOPA/BSZ + Mesdopetam 3 or 10 mg/kg
+Vehicle(saline)
LID SCORING
STOP
Baseline
-30 min
0
180 min
L-DOPA/BSZ
- Amantadine
-60 min
- Vehicle (DMSO)
- Pimavanserine
-90 min
Intact
Lesioned
Animal

Figure 1. A) Schematic of the experimental design. **B)** Drug administration procedures. **C)** Anatomical annotation of recorded structures following initial functional grouping. **D)** Summary of collected CT scans illustrating electrode recording positions in 114 different structures before functional grouping

References:
1) Cenci et al., *Cur. Prot. Neur.* 2007 2) Halje et al., *JN*, 2012

Dyskinesia scoring during electrophysiological recordings reveals antidyskinetic effects of amantadine and mesdopetam

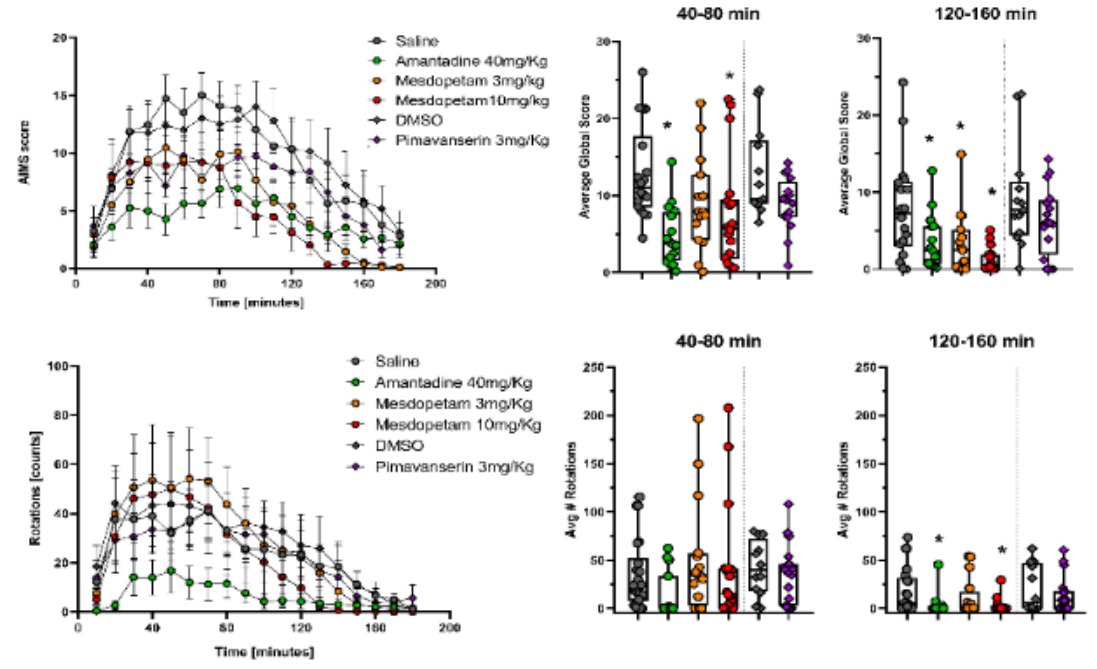
Saline
Amantadine 40mg/Kg
Mesdopetam 3mg/kg
Mesdopetam10mg/kg
DMSO
Pimavanserin 3mg/Kg
AIMS score
Time [minutes]
40-80 min
120-160 min
Average Global Score
Rotations [counts]
Avg # Rotations

Figure 2. A) Amantadine and mesdopetam reduced dyskinesia (AIMs scores) in both early and late phases of the recording period. **B)** Mesdopetam reduced the number of contralateral turns comparable to amantadine in the late phase (Kruskal-Wallis with Dunn post hoc test,*$p<0.05$. Graphs are box plots with 25-75% interquartile range).

Example spectrograms from MI illustrating drug-induced changes in narrowband gamma oscillations

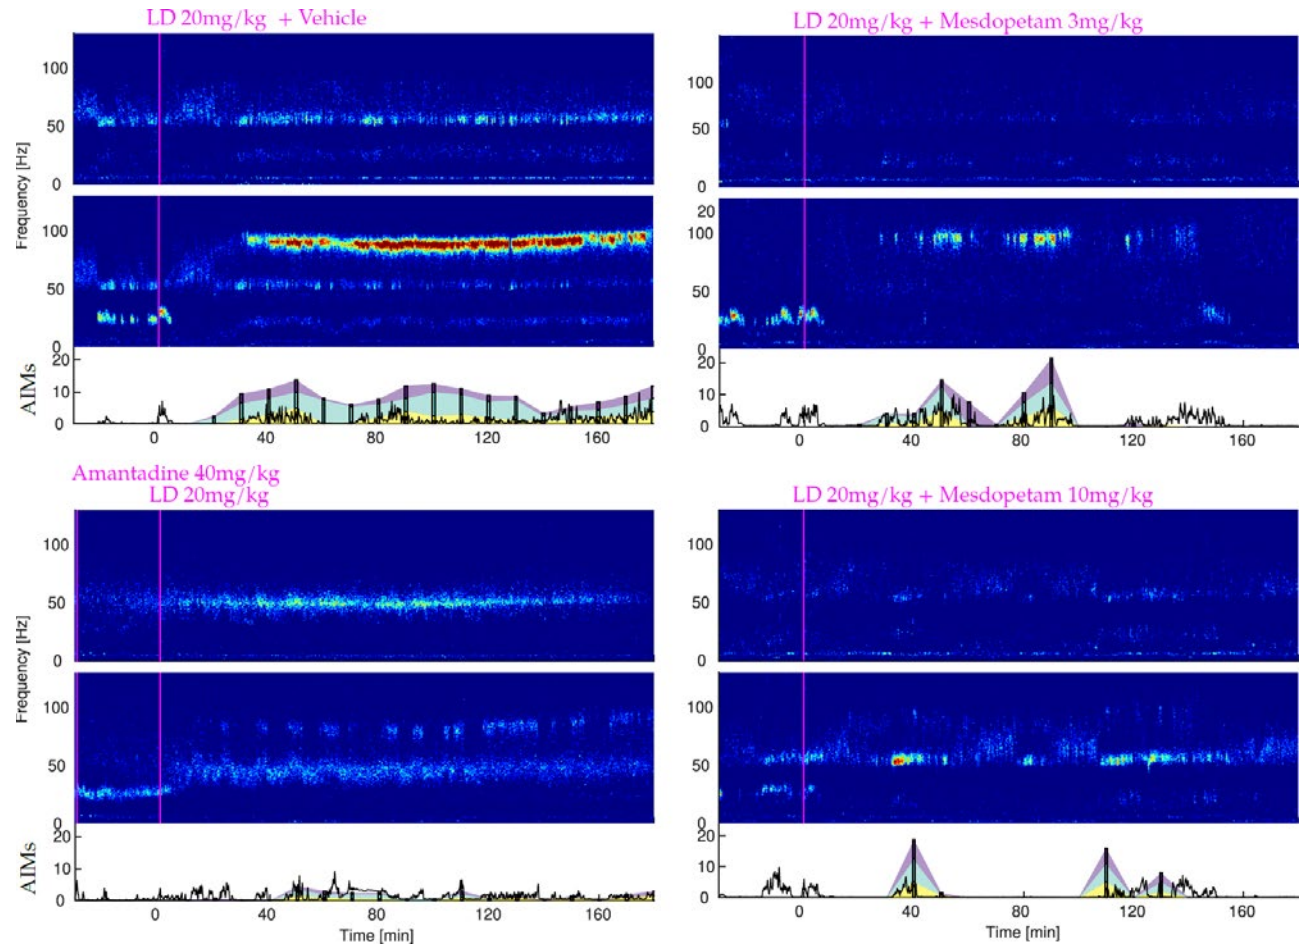
LD 20mg/kg + Vehicle
LD 20mg/kg + Mesdopetam 3mg/kg
Amantadine 40mg/kg
LD 20mg/kg
LD 20mg/kg + Mesdopetam 10mg/kg
Frequency [Hz]
AIMs
Time [min]

Figure 3. Amantadine, pimavanserin and mesdopetam suppress narrow-band gamma oscillations to a varying degree, strongly related to the reduction of AIMs. Veh: Vehicle; Pim: Pimavanserin; Mes3: Mesdopetam 3mg/kg; Mes10: Mesdopetam 10mg/kg

Reduction of oscillatory activity in the gamma-band is most pronounced in motor cortex

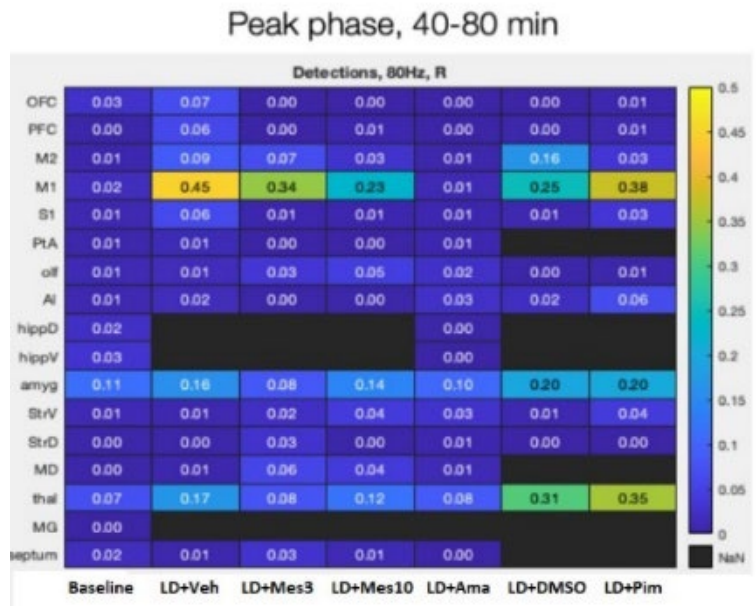
Peak phase, 40-80 min
Detections, 80Hz, R
Baseline
LD+Veh
LD+Mes3
LD+Mes10
LD+Ama
LD+DMSO
LD+Pim

Figure 4. Detection frequency of NBG in different brain area following treatments with amantadine, pimavanserin and mesdopetam. Veh: vehicle; Pim: Pimavanserine; Mes3: Mesdopetam 3mg; Mes10: Mesdopetam10mg

Drug-induced physiological states reveal differences in pharmacological mechanisms of action

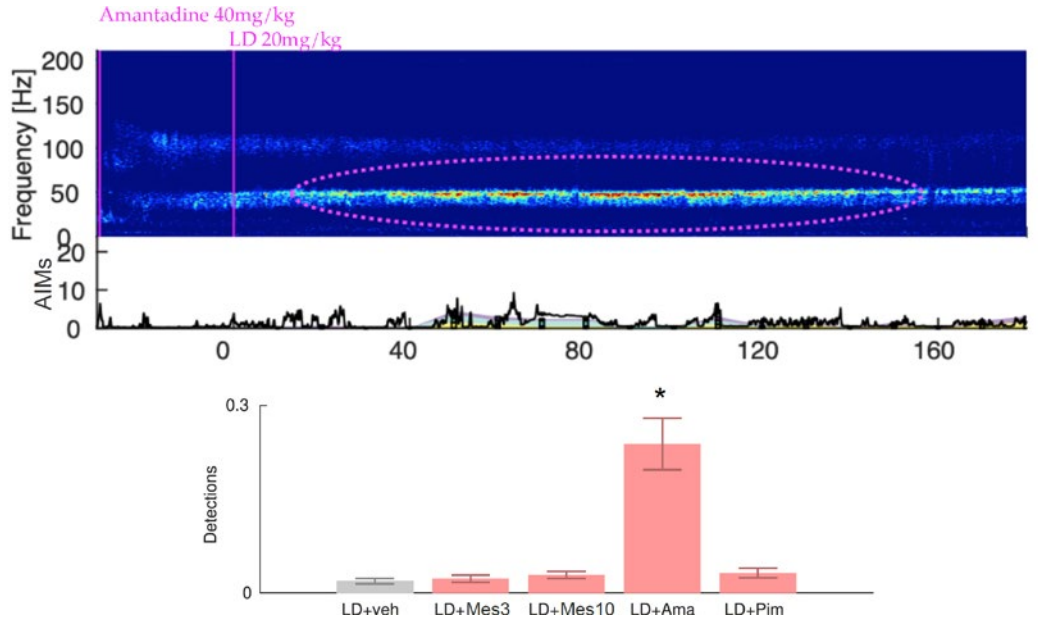
Amantadine 40mg/kg
LD 20mg/kg
Frequency [Hz]
AIMs
Detections
LD+veh
LD+Mes3
LD+Mes10
LD+Ama
LD+Pim

Figure 5. A) Example spectrogram from amygdala after treatment with amantadine revealed distinct oscillations in the gamma band that were not observed under any other condition. **B)** Frequency of detection of the amantadine-specific amygdala oscillations across all 11 rats

Conclusions and future directions

Mesdopetam and amantadine have comparable antidyskinetic effects in behavioral assessments of LID

Cortical narrow-band gamma oscillations are reduced by amantadine and mesdopetam, in a dose-dependent manner, while pimavanserin show only modest effects

A deeper analysis of spectral changes in the full frequency band will be needed to fully characterize differences in physiological treatment effects